#3616

AACR 102nd Annual Meeting 2011

Highly potent antibody-amanitin conjugates cause tumor-selective cell death

J. Anderl, C. Müller, W. Simon, C. Lutz, M. Kulke, C. Voss, B. Heckl-Östreicher, Roland Wehr
Heidelberg Pharma AG, Schriesheimer Str. 101, 68526 Ladenburg, Germany

INTRODUCTION

The therapeutic activity of a monoclonal antibody (mAb) to treat cancer is based on the activation of cellular responses triggered by Fc receptor binding or complement activation, the neutralization of target antigens or the modulation of tumor-related signaling pathways. These mechanisms do in most cases not eradicate tumor cells. Therefore, efforts were undertaken to expand the therapeutic efficacy of mAbs by covalent conjugation to small toxic compounds. These antibody-toxin conjugates or antibody-drug conjugates (ADC) bring together the specific target-binding capabilities of mAbs with the high cytotoxic potency of small molecular drugs in one single chemical entity. More than 20 years of reasearch on ADC revealed, that the best results were obtained with drugs to toxic to be used in free state. Thus todays most advanced ADC compounds depend on small and very toxic inhibitors of DNA replication, e.g. calicheamicins and duocarmycins, or modulators of tubulin polymerization, e.g. auristatin and maytansine analogues. As an example the ADC trastuzumab-DM1 (T-DM1; Genentech), a conjugate which consists of the HER2-binding antibody Herceptin® and the maytansine derivative DM1, shows promising results in clinical studies of HER2+ breast cancer.

In this study we present for the first time data on an ADC using the most potent and specific RNA pol II inhibitor α-amanitin using together with the mAb Herceptin. The Herceptin-amanitin conjugate Her-DSC-30.0134 has been evaluated in different *in vitro* and *in vivo* models of oncology and shown to possess potent antitumoral activity in HER2+ cells and tissues.

METHODS

Cell Lines: The human cancer cell lines SK-OV-3 (ovary), MDA-MB231 (breast) and NCI-N87 (stomach) were obtained from the ATCC and grown according to the suppliers instructions. JIMT-1 (breast) cells were obtained by the German Resource Center of Biological Material (DSMZ). HEK293 cells stably expressing the OATP1B3 transporter were established by standard protocols of transfection and selection.

Synthesis of Her-DSC-30.0134: α-Amanitin was covalently conjugated to lysine side chains of Herceptin by a proprietary, stable linker structure. Toxin payload of the resulting conjugate Her-DSC-30.0134 was adjusted to 3-4 amanitins per IgG molecule.

Antibody binding assay: 500 - 6.000 SK-OV-3, MDA-MB231 and NCI-N87 cells were plated in 96-well plates and incubated for 16h at 37°C and 5% CO_2. Cells were washed with PBS, pH 7.4 and fixed in 100% ice-cold MeOH for 10min at RT. Cells were incubated with 100µl 1µM Herceptin or Her-DSC-30.0134 in 5% BSA/PBS for 1h at RT. Detection of antibody binding was performed by a polyclonal rabbit anti-human IgG H&L POX antibody (Abcam) and luminol in a BMG Labtech multimode-reader.

Cell proliferation assay: Quantitative determination of DNA synthesis was performed by a chemoluminescent BrdU incorporation assay (Roche).

Mouse xenograft models: Six to 8-wk-old athymic female NMRI nude mice (nu/nu) were obtained from Janvier. SK-OV-3 cells (5.0 x 10^6) or JIMT-1 cells (5.0 x 10^6), respectively, were injected into the flank of each mouse for the development of a tumor. When tumors were well-established (150-250 mm^3), treatment was started. Tumor volume was calculated as $V = a \times b^2 \times 0.5$, where a is the length and b is the width.

www.heidelberg-pharma.com

RESULTS

Biological source and structure

The small molecule compound α-amanitin (MW = 919) is the main component of the amatoxins, bicyclic octapeptides produced by basidiomycetes of the genus *Amanita*, e.g. the Green Deathcap mushroom (*Amanita phalloides*) (Fig. 1).

(a) (b)

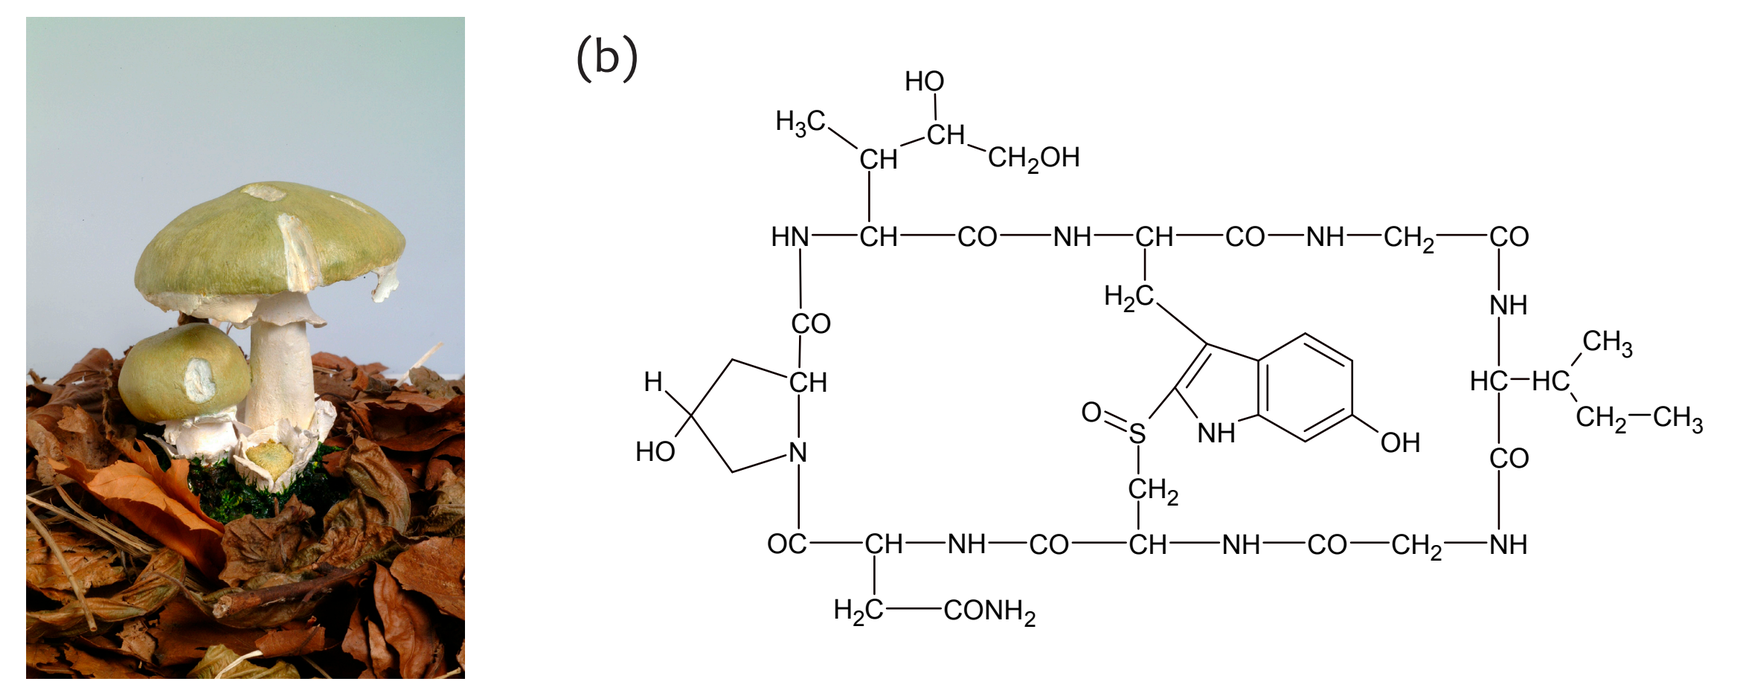

Figure 1: (a) Green Deathcap mushroom (*Amanita phalloides*). (b) α-Amanitin.

Low toxicity on cells expressing amanitin liver transporter

While widely used as a tool in molecular biology, only a few attempts have been made to employ α-amanitin as a therapeutic agent in oncology. The reason for this may be the high liver toxicity, caused by the transport of the membrane-impermeable α-amanitin into mature hepatocytes by the organic-anion transporter 1B3 (OATB1B3) present on hepatocytes, but not on other cell types[1]. After covalent conjugation to mAbs the small molecule α-amanitin is no longer a substrate for OATP1B3 mediated transport, as shown for Her-DSC-30.0134 using HEK293-OATP1B3 cells (Fig. 2). While HEK293-OATP1B3 cells show a pronounced sensitivity against α-amanitin compared to non-transfected HEK293 cells, Her-DSC-30.0134 conjugate was only weakly toxic, independent of the transporter status of cells. This suggests reduced liver toxcity and enhanced tolerability of antibody-conjugated α-amanitin compared to the unconjugated natural product.

(a) (b)

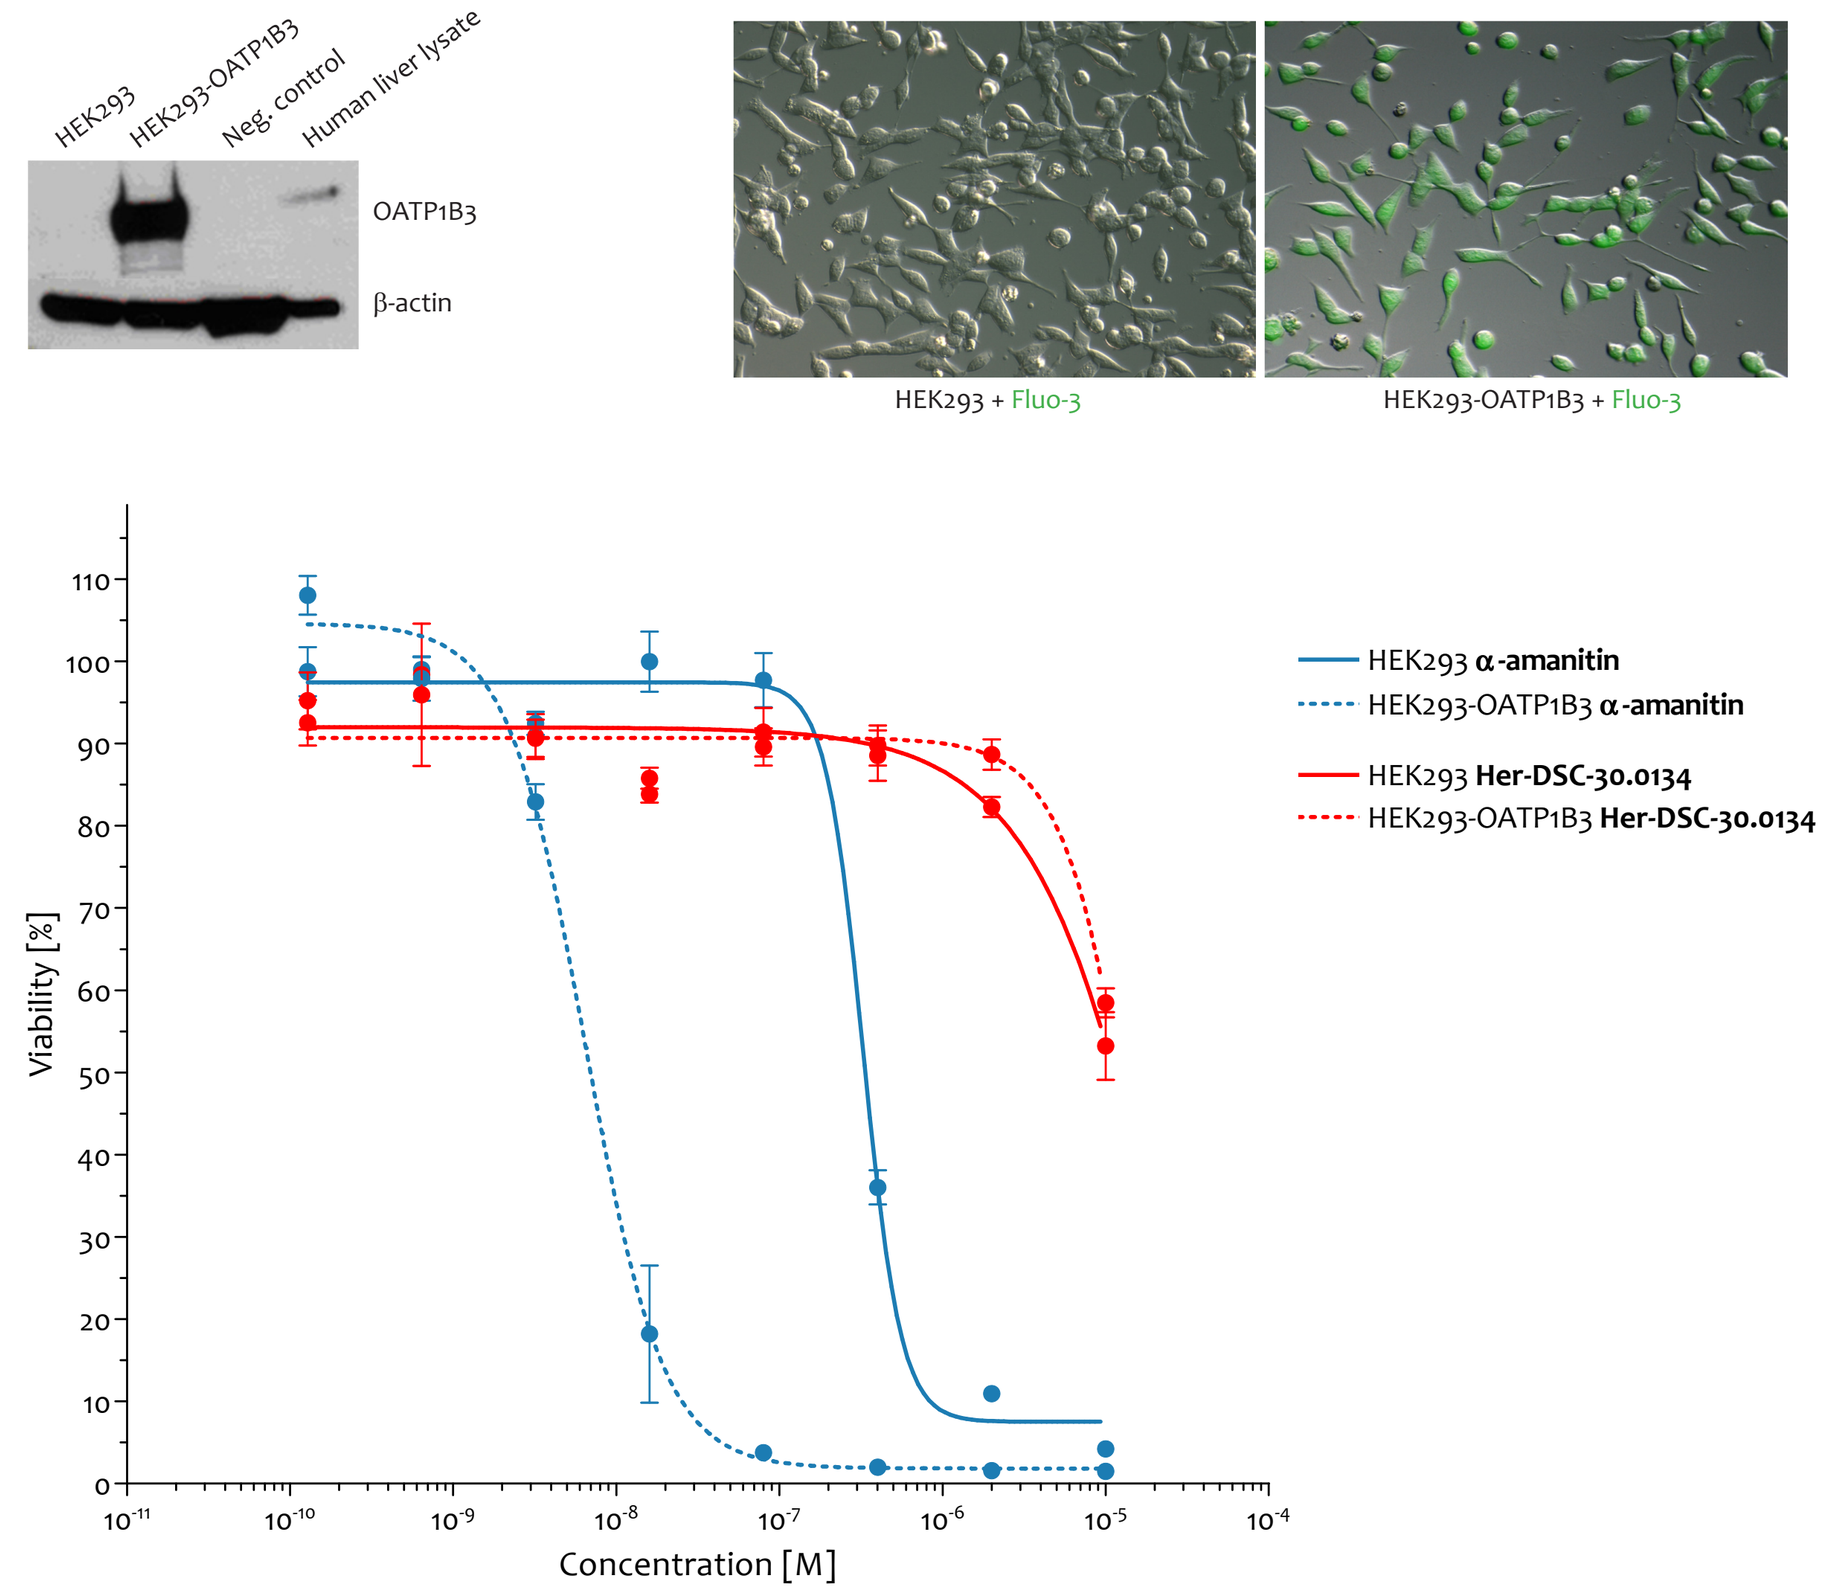
HEK293
HEK293-OATP1B3
Neg. control
Human liver lysate
OATP1B3
β-actin
HEK293 + Fluo-3
HEK293-OATP1B3 + Fluo-3
Viability [%]
Concentration [M]
HEK293 α-amanitin
HEK293-OATP1B3 α-amanitin
HEK293 Her-DSC-30.0134
HEK293-OATP1B3 Her-DSC-30.0134

Figure 2: (a) Western Blot analysis and Fluo-3 uptake assay demonstrating the functionality of OATB1B3 in transfected HEK293 cells. (b) BrdU cell proliferation assay with HEK293 and HEK293-OATP1B3 cells after 72h incubation with different concentrations of α-amanitin or Her-DSC-30.0134.

Cell binding and cytotoxicity *in vitro*

Binding of Herceptin and Her-DSC-30.0134 was compared for SK-OV-3, NCI-N87 and MDA-MB231 cells expressing on the cell surface high, medium and low levels of HER2, respectively. Data revealed, that binding of Herceptin correlates with antigen surface expression on the different cell lines (Fig. 3). Drug payload of 3-4 amanitins per IgG does not effect antigen binding of Herceptin using the proprietary linker structure of Her-DSC-30.0134.

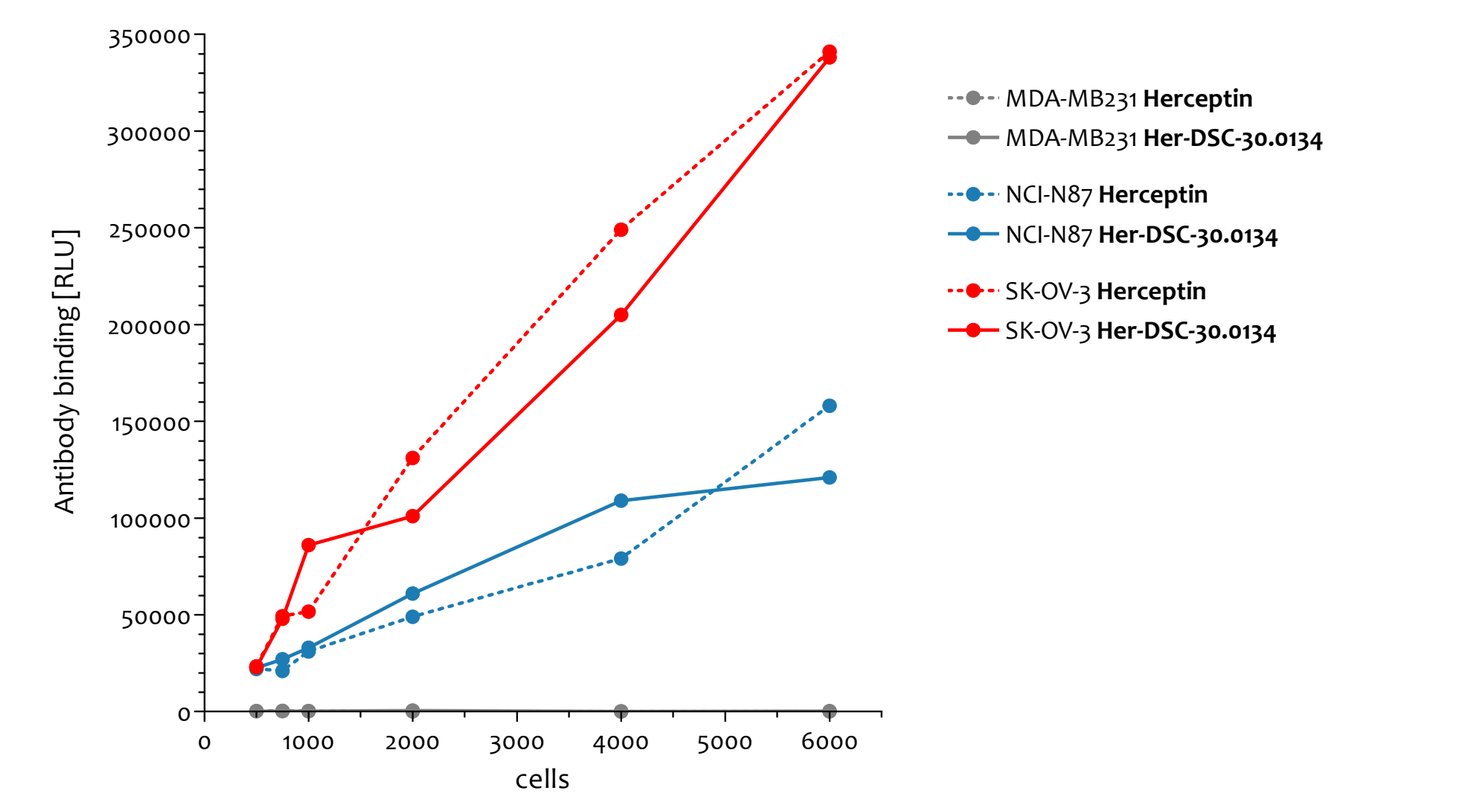
Antibody binding [RLU]
cells
MDA-MB231 Herceptin
MDA-MB231 Her-DSC-30.0134
NCI-N87 Herceptin
NCI-N87 Her-DSC-30.0134
SK-OV-3 Herceptin
SK-OV-3 Her-DSC-30.0134

Figure 3: Cell surface binding of Herceptin and Her-DSC-30.0134 on cells with varying HER2 surface expression.

Cytotoxic activity of Her-DSC-30.0134 vs. the toxin-free Herceptin has been confirmed for SK-OV-3, NCI-N87 and MDA-MB231 cells by a BrdU-incorporation assay (Fig. 4). The cytotoxic activity of Her-DSC-30.0134 correlates with antigen expression and ADC binding, showing high cytotoxic activity with SK-OV-3 cells, medium activity with NCI-N87 cells and no activity within the used concentration range with HER2 lacking MDA-MB231 cells. Unconjugated Herceptin is not toxic up to a concentration range 10^3-10^4 times higher for all three cell lines. Her-DSC-30.0134 is approx. 50.000 to 600.000 times more toxic than unconjugated α-amanitin, which shows only moderate cytotoxicity on OATP1B3-negative cells because of its low membrane permeability (Table 1).

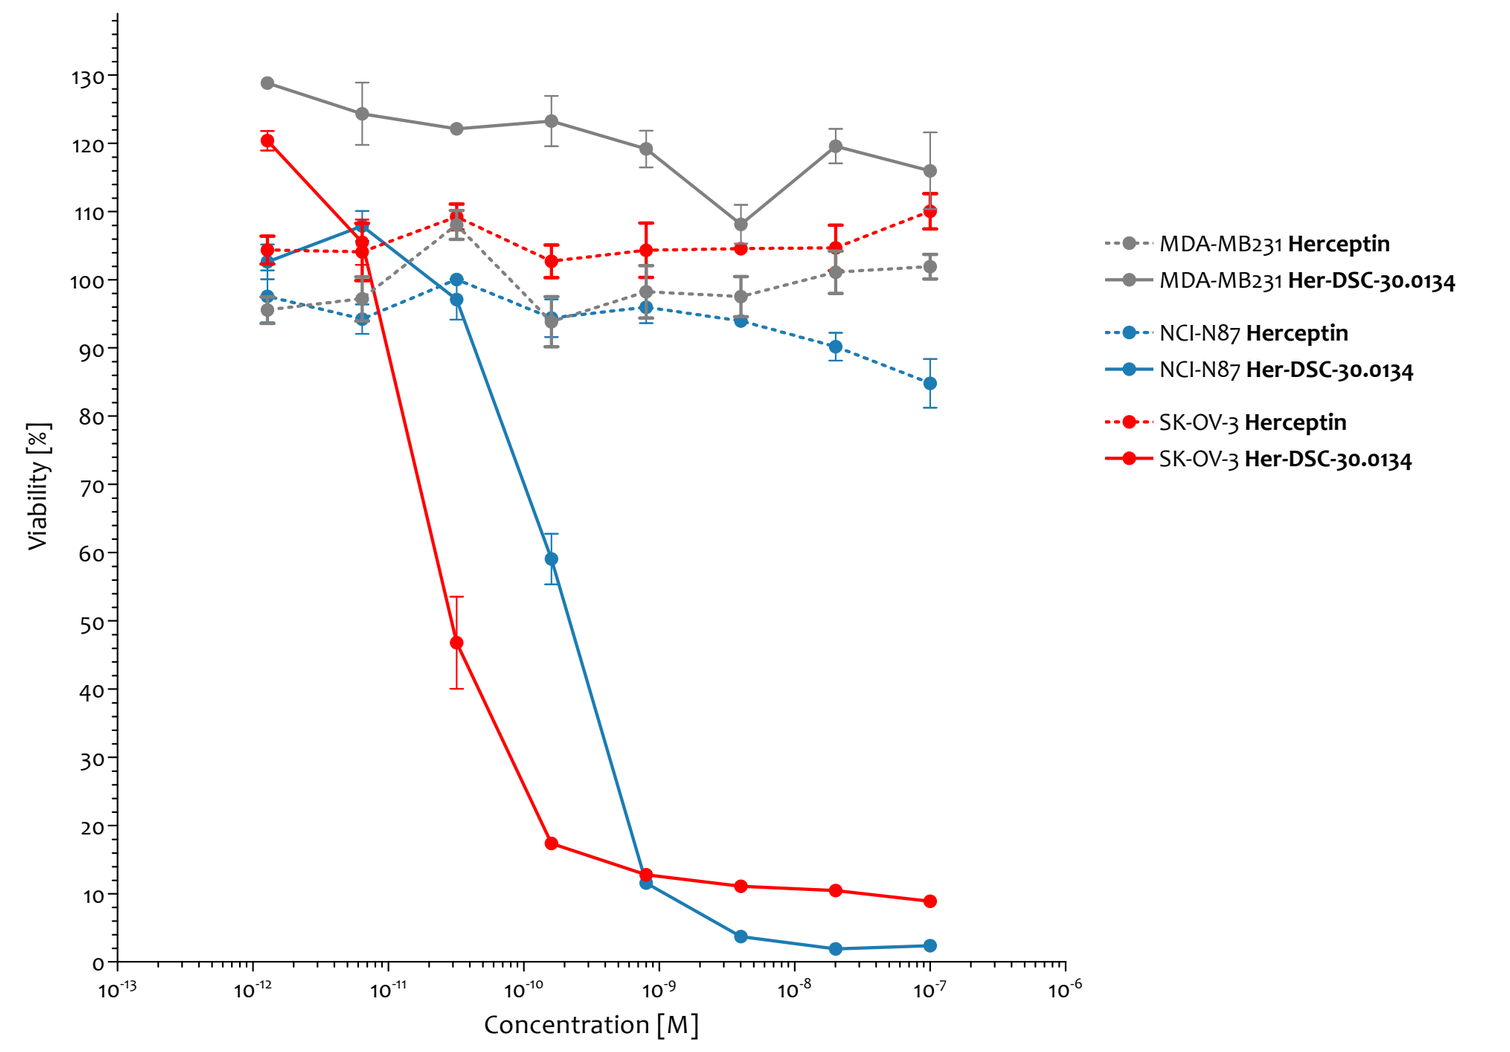
Viability [%]
Concentration [M]
MDA-MB231 Herceptin
MDA-MB231 Her-DSC-30.0134
NCI-N87 Herceptin
NCI-N87 Her-DSC-30.0134
SK-OV-3 Herceptin
SK-OV-3 Her-DSC-30.0134

Figure 4: BrdU cell proliferation assay with tumor cells after 72h incubation with different concentrations of Herceptin or Her-DSC-30.0134.

Cell line	Her-DSC-30.0134 EC50 [M]	Toxicity compared to α-amanitin
SK-OV-3	1.96×10^{-11}	510.000-fold
SK-BR-3	8.2×10^{-12}	590.000-fold
NCI-N87	1.82×10^{-10}	49.000-fold
MDA-MB231	$>1.0 \times 10^{-7}$	nd

Table 1: EC50 concentrations of Her-DSC-30.0134 in HER2+ cell lines SK-OV-3, SK-BR-3 and NCI-N87 and in the HER2- cell line MDA-MB231.

Antitumoral activity in HER2+ mouse xenograft models

Antitumoral activity of Her-DSC-30.0134 was demonstrated in a Herceptin-sensitive SK-OV-3 s.c. mouse xenograft model. A single dose of 50µg/kg Her-DSC-30.0134, with respect to amanitin, showed complete tumor remission in 7/7 animals without recurrence within >80 days (Fig. 5a). The applied doses of Her-DSC-30.0134 did not effect body weight (Fig. 5b) or liver function, as monitored by plasma ALT values. Moreover, Her-DSC-30.0134 showed curative activity in a xenograft model using Herceptin and lapatinib (Tykerb®) resistant JIMT-1 cells, isolated from the pleural effusion of a HER2+ breast cancer patient[2]. 7/7 animals were tumor-free 18 days after a single i.v. injection of 150µg/kg and even of 30µg/kg Her-DSC-30.0134. Unconjugated Herceptin showed only minor antitumoral activity despite multiple applications of higher doses. Doxil® therapy showed moderate actitivy at a dose leading to >20% body weight reduction of animals.

(a) (b)

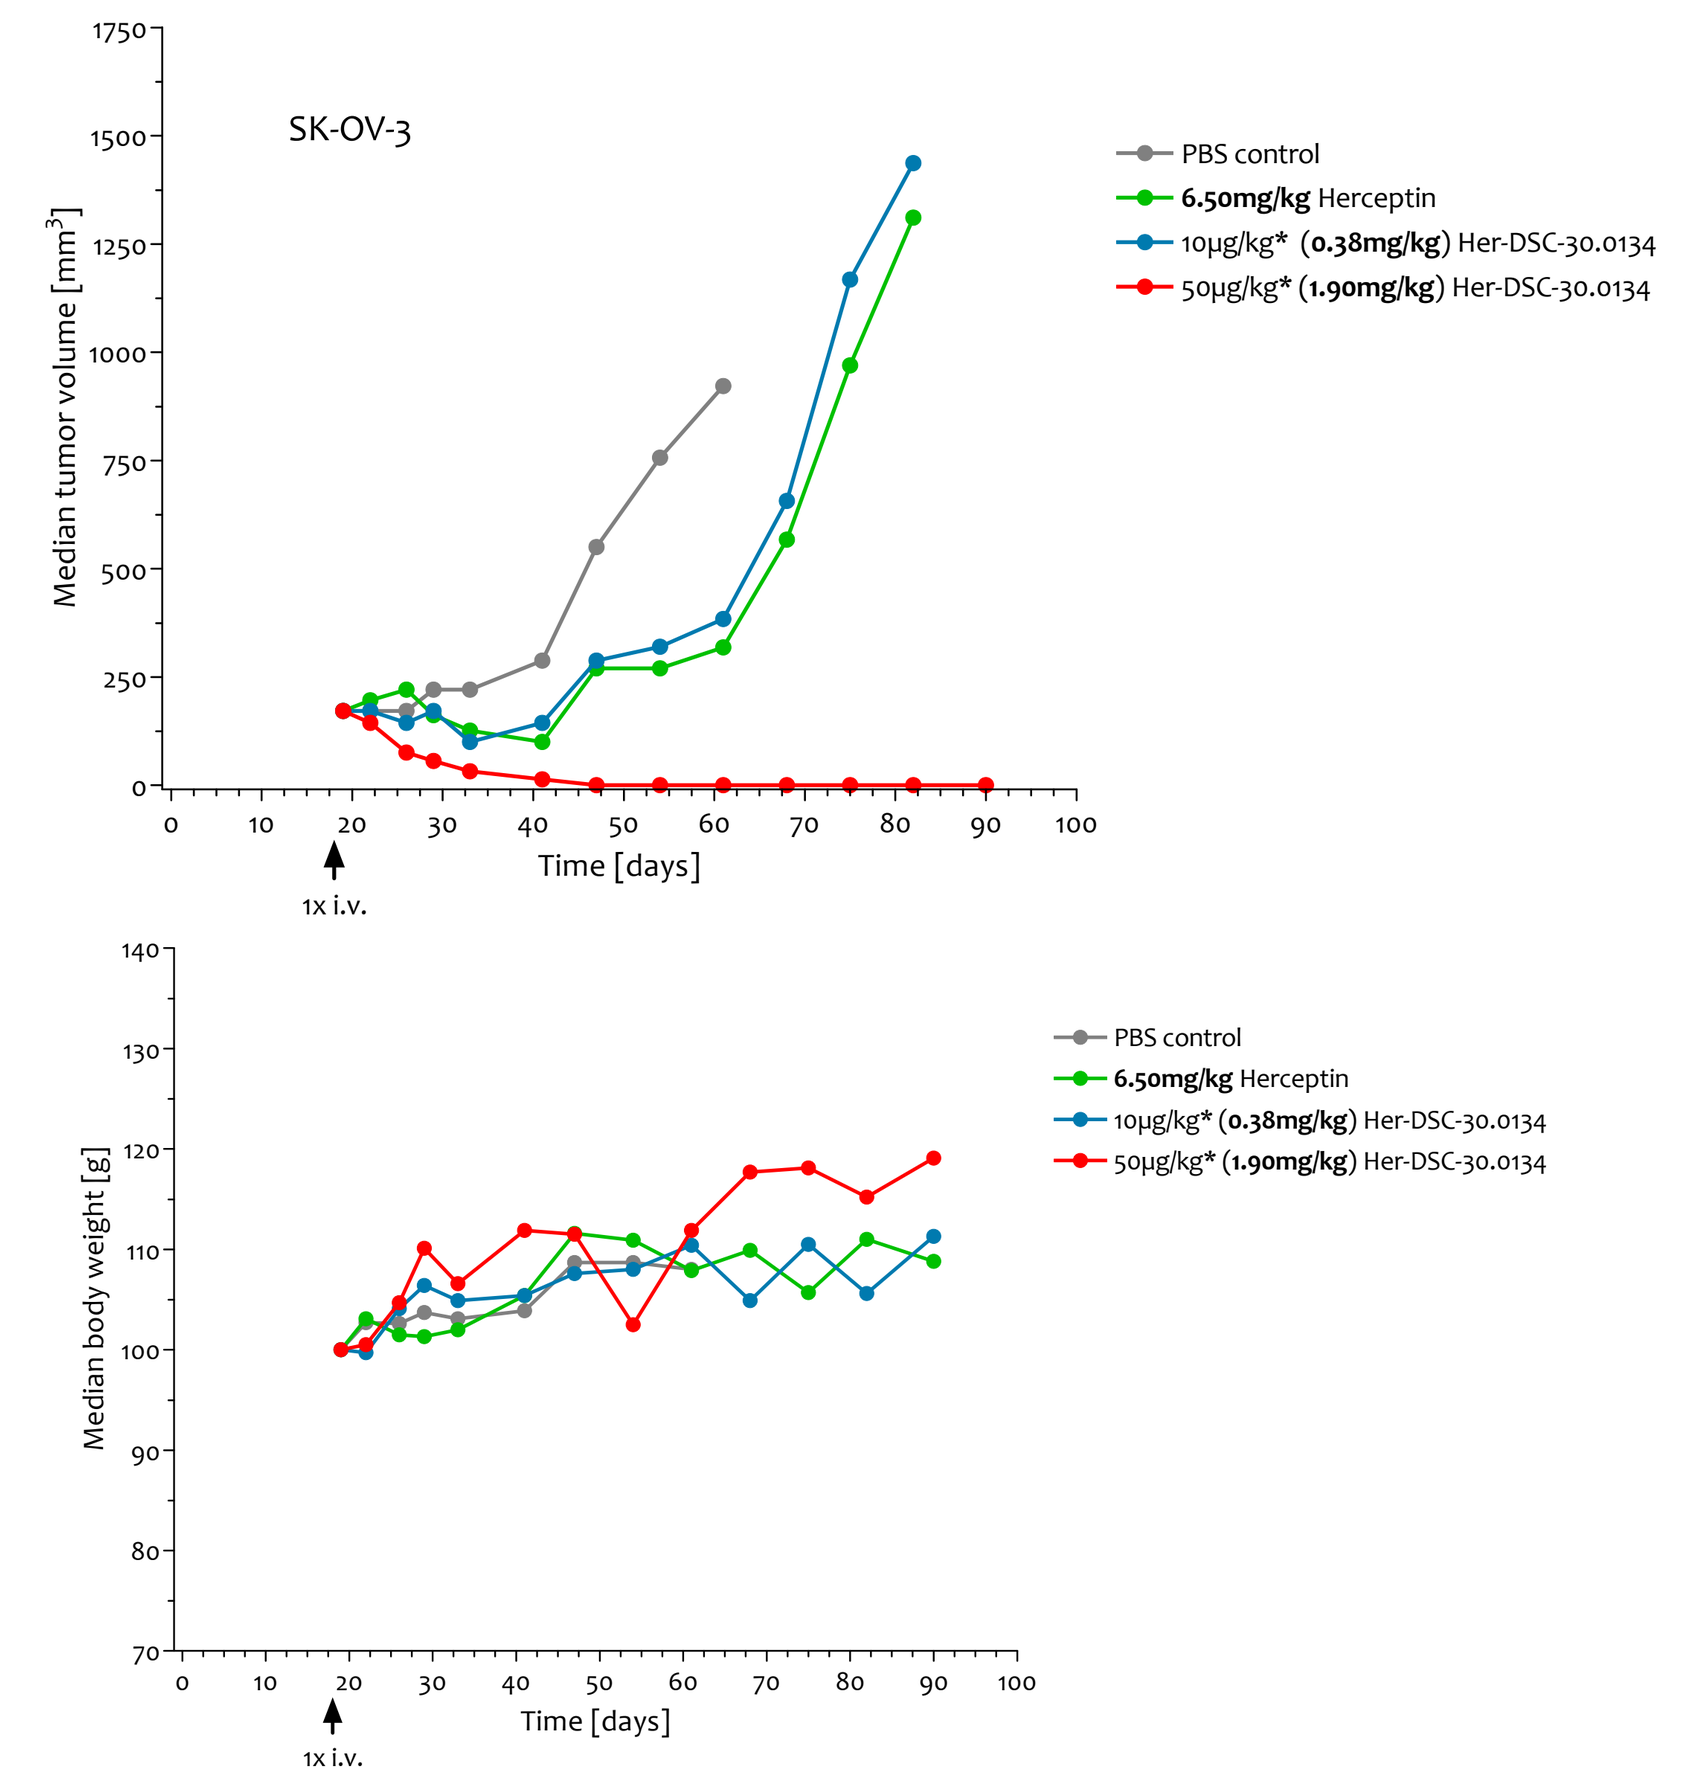
SK-OV-3
Median tumor volume [mm³]
Median body weight [g]
Time [days]
1x i.v.
PBS control
6.50mg/kg Herceptin
10µg/kg* (0.38mg/kg) Her-DSC-30.0134
50µg/kg* (1.90mg/kg) Her-DSC-30.0134

Figure 5: (a) SK-OV-3 mouse xenograft model using 1x i.v. injection of Herceptin or Her-DSC-30.0134. (b) Body weight change of mice during SK-OV-3 experiment. (*doses with respect to amanitin)

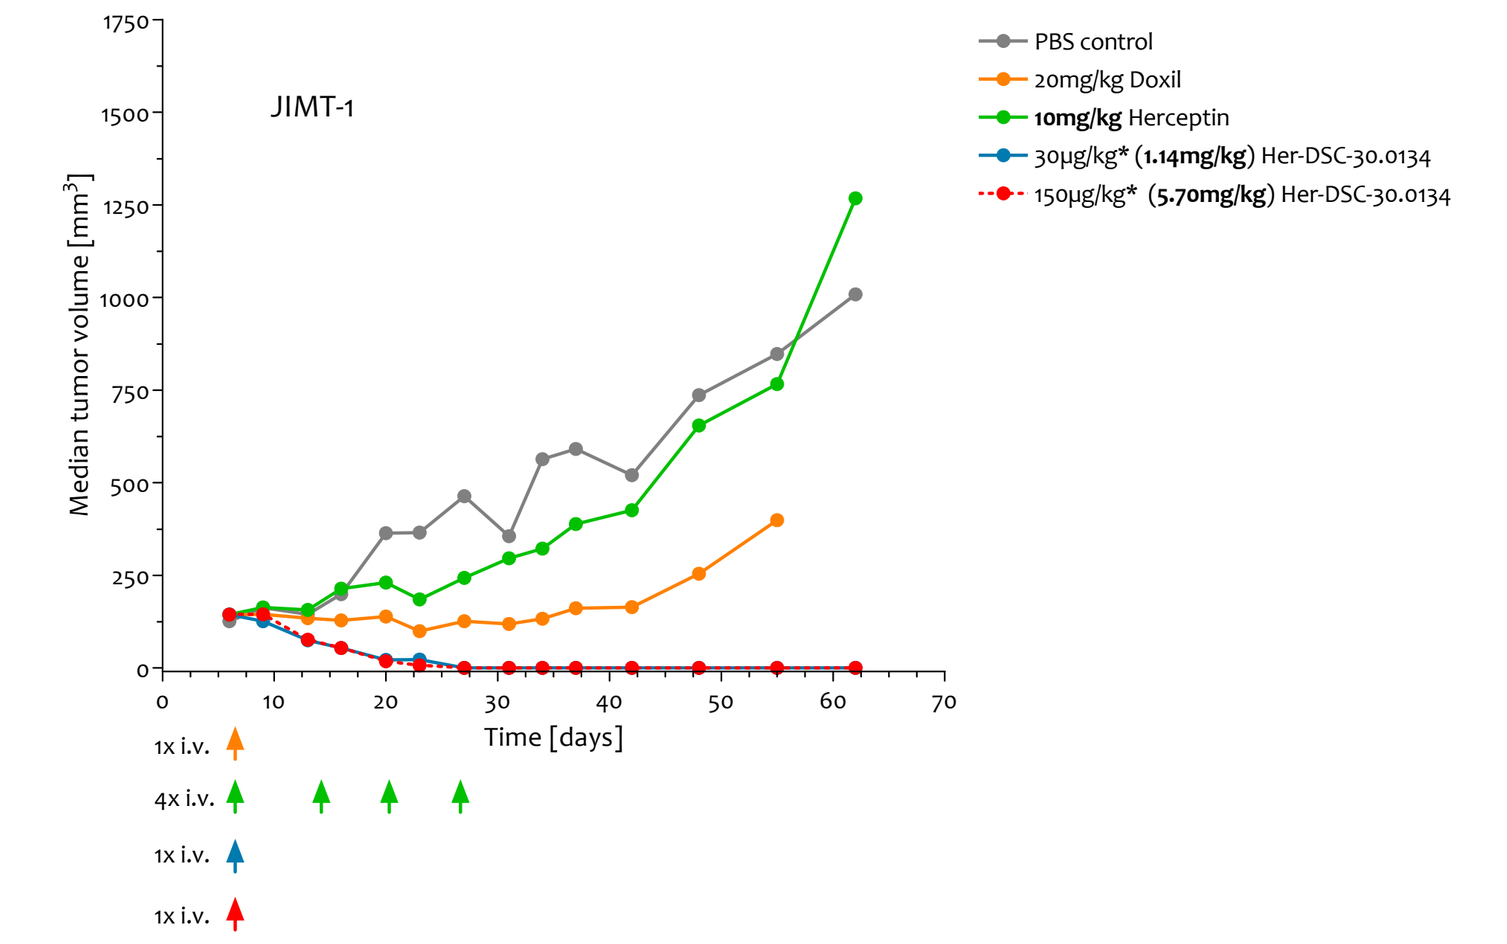
JIMT-1
Median tumor volume [mm³]
Time [days]
1x i.v.
4x i.v.
1x i.v.
1x i.v.
PBS control
20mg/kg Doxil
10mg/kg Herceptin
30µg/kg* (1.14mg/kg) Her-DSC-30.0134
150µg/kg* (5.70mg/kg) Her-DSC-30.0134

Figure 6: JIMT-1 mouse xenograft model using 4x i.v. injection of Herceptin or 1x i.v. injection of Her-DSC-30.0134 and Doxil. (*doses with respect to amanitin)

CONCLUSIONS

The presented data demonstrate for the first time exceptional antitumoral activity by *in vitro* and *in vivo* models of oncology using a direct inhibitor of RNA pol II. Antibody-conjugation of the small molecule compound α-amanitin prevents liver-uptake by OATP1B3 transport and hence allows safe application of effective doses of Her-DSC-30.0134 ADC in animal models. Stable linker structures have been found in order to minimize release of toxin, thereby reducing the risk of toxicity to OATP1B3-expressing hepatotcytes. Furthermore, Herceptin-conjugation not only targets to tumor cells, but also leads to accumulation of the toxin in the tumor cells. Stalemate of transcription by directly inhibiting the transcribing enzyme RNA pol II resulted in eradication of cancer cells at picomolar concentrations. Finally, using *in vivo* models, Her-DSC-30.0134 demonstrated high antitumoral activity in Herceptin-sensitive and even Herceptin and lapatinib resistant tumors.

Other qualities that argue for α-amanitin as drug in ADCs:

- α-Amanitin differs from other toxins used in todays ADC therapeutics by its unique intracellular target.
- Amanitin-based ADCs like Her-DSC-30.0134 are expected not only to to destroy proliferating cells but also cells with low proliferation rate, e.g. tumor stem cells.
- The toxin is characterized by its high systemic and proteolytic stability.
- α-Amanitin is insensitive to Pgp-mediated resistance mechanisms of cancer (data not shown).

The positive findings of these initial experiments encourage Heidelberg Pharma to use α-amanitin as a basis for an innovative ADC platform technology.

REFERENCES

(1) Letschert K, Faulstich H, Keppler D. Molecular characterization and inhibition of amanitin uptake into human hepatocytes. *Toxicol Sci.* 2006; 91(1):140-9

(2) Tanner M, Kapanen AI, Junttila T, Raheem O, Grenman S, Elo J, Elenius K, Isola J. Characterization of a novel cell line established from a patient with Herceptin-resistant breast cancer. *Mol Canc Ther.* 2004; 3(12):1585-92

CONTACT

Dr. Marcel Linssen

CBO (Executive Vice President Corporate Development & Marketing)
Heidelberg Pharma AG
Schriesheimer Str. 101
68526 Ladenburg / Germany

Tel. +49-6203-1009 40
Email: m.linssen@hdpharma.com